P.1015

Cariprazine's efficacy in residual patients with schizophrenia: a post-hoc analysis in a chronic patient population

Zs. B. Dombi[1], Á. Barabássy[1], R. Csehi[1], B. Sebe[1], K. Acsai[1], I. Laszlovszky[1], B. Szatmári[1], J. Harsányi[1], G. Németh[1]

[1]*Richter Gedeon Plc., Medical Division, Budapest, Hungary*

INTRODUCTION

According to the International Classification of Diseases 10th edition (ICD-10), residual schizophrenia (F20.5) is defined as a chronic stage of the disorder where patients follow a clear progression from early to later stages of the illness and are experiencing long-term, but not necessarily irreversible, negative symptoms such as blunted affect, as well as cognitive and functional difficulties (1).The pharmacotherapy of residual schizophrenia is quite challenging to clinicians not only because of the nature of the symptoms but due to the fact that older patients are more susceptible to adverse events associated with antipsychotic medication as well (2). Cariprazine, a novel D3-D2 partial agonist has been proven to be safe and effective in the treatment of predominant negative symptoms associated with schizophrenia (3), however its ability to alleviate the symptoms of residual patients has not been specifically assessed yet.

STUDY OBJECTIVE

The primary aim of the present post-hoc analysis is to assess the efficacy of cariprazine in treating patients with residual schizophrenia measured by the Factor Score for Negative Symptoms of the Positive and Negative Syndrome Scale (PANSS-FSNS) and the Personal and Social Performance Scale (PSP) compared to risperidone.

METHODS

This was a 26-week, international, randomized, double-blind, active-controlled, fixed-flexible-dose trial with schizophrenia patients experiencing predominantly negative symptoms as defined by having a score of 24 or higher on the PANSS-FSNS with no pseudo-specific factors such as high levels of positive symptoms. Enrolled patients were randomized to a daily dose of 4.5 mg cariprazine (n=227) with a dose range of 3.0-6.0 mg/day or 4.0 mg risperidone (n=229) with a dose range of 3.0-6.0 mg/day. Patients with an ICD-10 code of F20.5 were analysed post-hoc.

RESULTS

A total of 23 patients were classified as having residual schizophrenia from the cariprazine (CAR) and 12 in the risperidone (RIS) group. Baseline characteristics of the patients are summarised in Table 1. The mean age of patients was 44 for CAR and 40 for RIS-treated patients. In average, the duration of schizophrenia was 14 years in the CAR and 15 years in the RIS group. The baseline PANSS-FSNS and PSP scores in the CAR group were 27.5 and 49.6, respectively. In the RIS group the baseline PANSS-FSNS was 26.9, while the PSP was 52.7.

Table 1: Baseline demographic and outcome scores

	Cariprazine 4.5 mg/day	Risperidone 4.0 mg/day
Demographics		
Number of patients, n	23	12
Male, n (%)	16 (69.6)	5 (41.7)
Mean Age, n (SD)	43.6 (10.4)	40.1 (10.6)
Mean duration of schizophrenia, n (SD)	14.1 (9.2)	15.2 (9.3)
Baseline Scores		
Baseline PANSS-FSNS, mean (SD)	27.5 (2.2)	26.9 (2.2)
Baseline PSP, mean (SD)	49.6 (10.2)	52.7 (6.2)

The mean change from baseline in patients with residual schizophrenia in the cariprazine arm was -9.6 on the PANSS-FSNS scale, while -7.9 in the risperidone arm. Importantly, these results are quite similar to the non-residual population where the mean change from baseline in the cariprazine and risperidone group was -9.6 and -7.8, respectively. Due to low residual schizophrenia patient numbers no significance could be measured.

CONCLUSIONS

Based on the results of the post-hoc analysis it seems that cariprazine might be a good treatment option for patients with residual schizophrenia. Nonetheless, due to small sample size further studies are needed to confirm the results.

Figure 1: Mean change from baseline in PANSS-FSNS

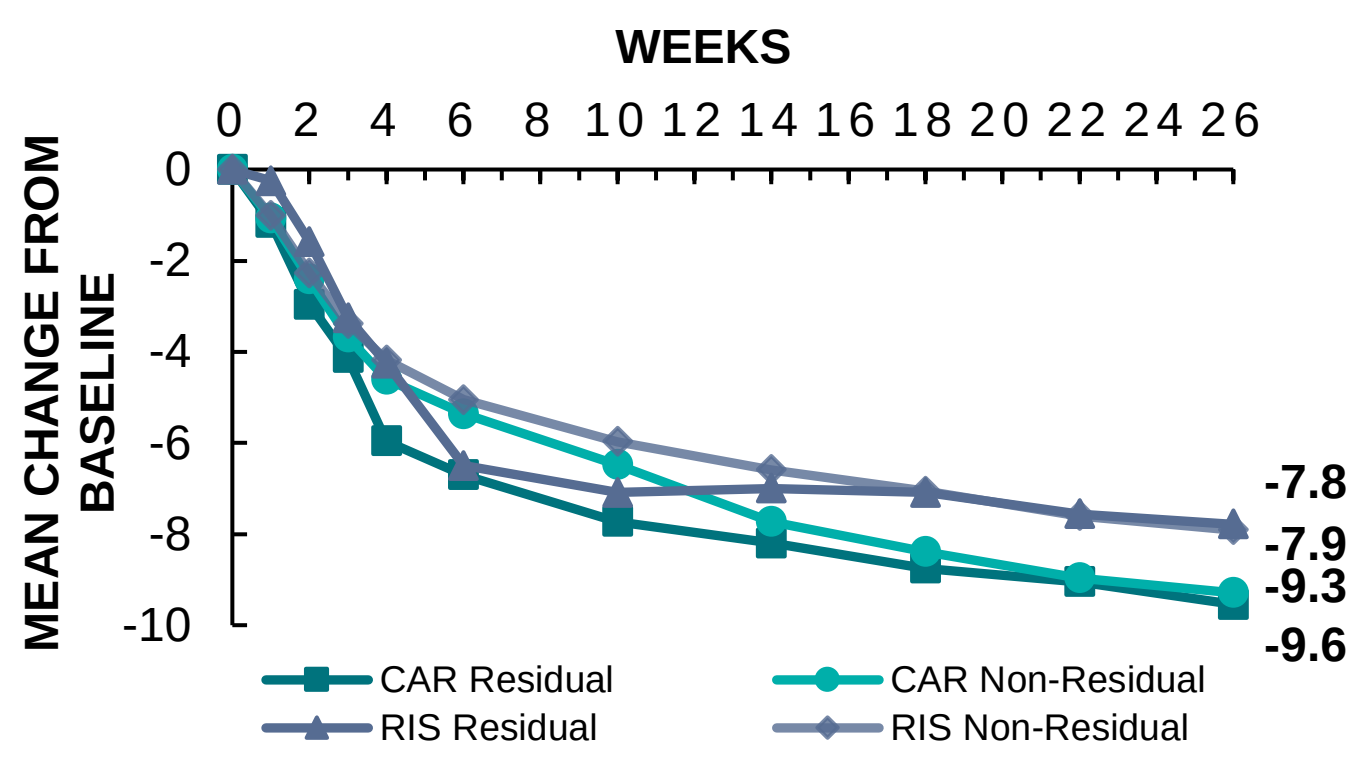
WEEKS
0 2 4 6 8 10 12 14 16 18 20 22 24 26
0
-2
-4
-6
-8
-10
MEAN CHANGE FROM BASELINE
-7.8
-7.9
-9.3
-9.6
CAR Residual
CAR Non-Residual
RIS Residual
RIS Non-Residual

When analysing the mean change from baseline on the PSP scale, patients with residual schizophrenia receiving cariprazine showed an average improvement of 13.6 points by the end of the 26th week, while the same number was 15.3 for non-residual patients. In contrast, patients on risperidone treatment had smaller change from baseline; -10.9 in residual and 11.5 in non-residual population.

Figure 2: Mean change from baseline in PSP

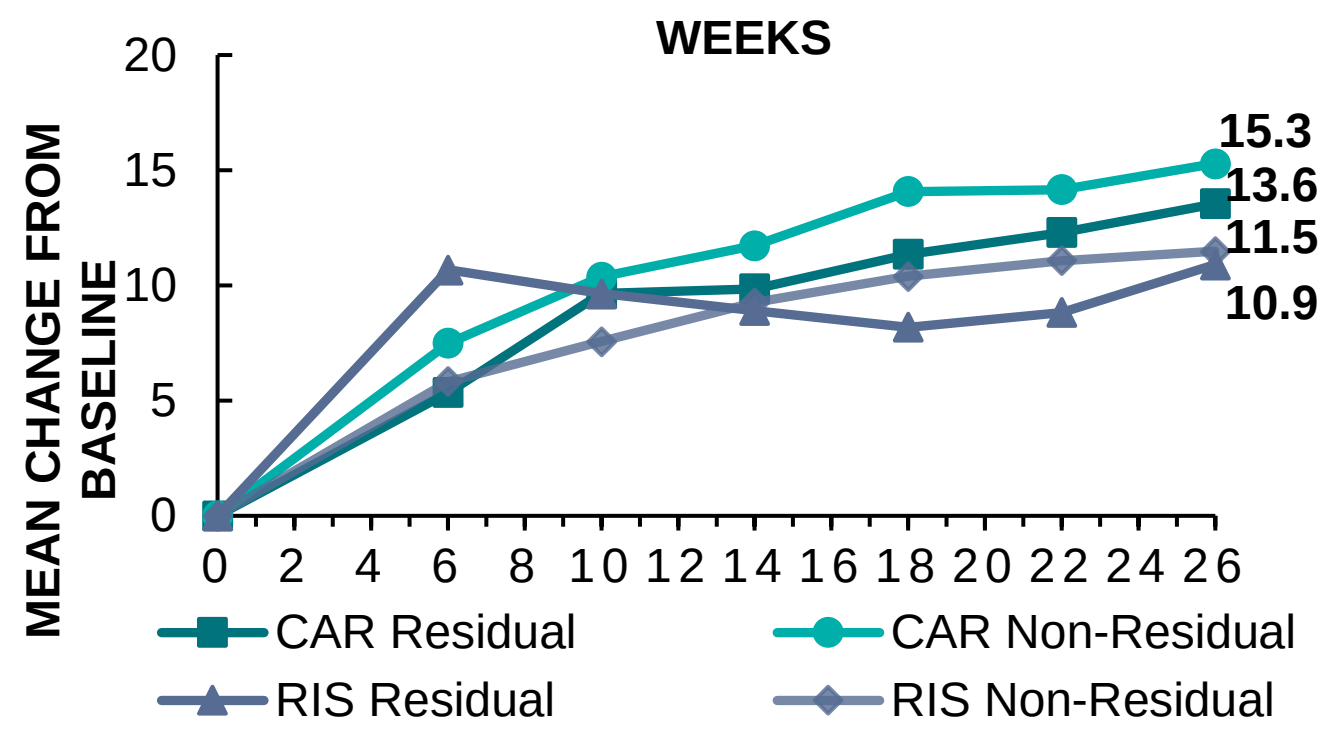
WEEKS
20
15
10
5
0
MEAN CHANGE FROM BASELINE
15.3
13.6
11.5
10.9
0 2 4 6 8 10 12 14 16 18 20 22 24 26
CAR Residual
CAR Non-Residual
RIS Residual
RIS Non-Residual

REFERENCES

1. World Health Organization. The ICD-10 Classification of Mental and Behavioural Disorders. Diagnostic criteria for research. Int Classif. 1993;
2. Khan A, Kalia R, Ide G, Ghavami M. What Are Realistic Treatment Goals? Lingering Symptoms Require You to Evaluate Pharmacotherapy and Offer Psychosocial Interventions. Curr Psychiatr. 2017;16(3):35–40.
3. Németh G, Laszlovszky I, Czobor P, Szalai E, Szatmári B, Harsányi J, et al. Cariprazine versus risperidone monotherapy for treatment of predominant negative symptoms in patients with schizophrenia: a randomised, double-blind, controlled trial. Lancet. 2017;389(10074):1103–13.

DISCLOSURES & FUNDING STATEMENT

Studies were funded by Gedeon Richter Plc. Dombi, Dr. Barabássy, Csehi, Dr. Sebe, Acsai, Dr. Laszlovszky, Dr. Szatmári, Dr. Harsányi and Dr. Németh are employees of Gedeon Richter Plc.

Presented at the 34th European College of Neuropsychopharmacology Congress, October 2-5, 2021; Hybrid